Analysis of Time to Next Treatment in Melflufen and Dexamethasone-Treated Patients With Relapsed/Refractory Multiple Myeloma

Poster 8043

Sara Bringhen, MD[1]; Paul G. Richardson, MD[2]; Peter Voorhees, MD[3]; Torben Plesner, MD[4]; Ulf-Henrik Mellqvist, MD[5]; Jeffrey A. Zonder, MD[6]; Brandi Reeves, MD[7]; Stojan Zavisic, BSc[8]; Johan Harmenberg, MD[8]; Jakob Obermüller, MSc[8]; and Pieter Sonneveld, MD[9]

[1]Division of Hematology University of Torino, Torino, Italy; [2]Dana-Farber Cancer Institute, Harvard Medical School, Boston, MA, USA; [3]Levine Cancer Institute, Carolinas HealthCare System, Charlotte, NC, USA; [4]Department of Hematology, Vejle Hospital, Vejle, Denmark; [5]Borås Hospital, Borås, Sweden; [6]Karmanos Cancer Institute, Wayne State University, Detroit, MI, USA; [7]Lineberger Comprehensive Cancer Center, University of North Carolina, Chapel Hill, NC, USA; [8]Oncopeptides AB, Stockholm, Sweden; [9]Erasmus Medical Center Rotterdam, Rotterdam, The Netherlands

BACKGROUND

- In a patient with progressing relapsed/refractory multiple myeloma (RRMM), an important clinical objective is to achieve durable disease stabilization (stable disease [SD] or better), especially in patients with moderate to low disease burden. With each relapse, a patient's prognosis worsens, and time to the next relapse decreases[1,2]
- Time to next treatment (TTNT) reflects time of disease stabilization in patients with RRMM and may be a relevant parameter of clinical benefit for patients, clinicians, and health economists[1,3]
- TTNT is an important parameter to help assess cost of care and is used in real-world evidence (RWE) to assist treatment decisions and support economic reimbursement modeling[1,3,4]
 - Longer TTNT has previously been associated with lower costs in multiple myeloma[1,4]
- Melflufen is a lipophilic peptide-conjugated alkylator that rapidly delivers a highly cytotoxic payload into myeloma cells through peptidase activity (**Figure 1**)[5-9]
- O-12-M1 is a phase 1/2 study with melflufen plus dexamethasone in 62 patients with RRMM who had ≥2 prior lines of therapy, prior exposure to at least an IMiD and a proteasome inhibitor, and disease progression on last line of therapy. Final study results were presented at ASH 2017[10]
- In O-12-M1, cycle lengths of 21 and 28 days were assessed, and a cycle length of 28 days was determined to be optimal with regard to dose intensity and safety profile[10]
- The phase 2 part of O-12-M1 showed encouraging activity[10]
 - Median 4-5 prior lines of therapy, 44% high-risk cytogenetics, 67% double refractory, 44% pomalidomide refractory, 60% ISS stage 2-3
 - Overall response rate (ORR; ≥partial response), 31%; median duration of response (DOR), 8.4 months
 - Clinical benefit rate (CBR; ≥minimal response), 49%
 - Median progression-free survival (PFS), 5.7 months
 - Median overall survival (OS), 20.7 months
 - The most common grade 3 and 4 treatment-emergent adverse events (TEAEs) were hematologic. Grade 3 and 4 nonhematologic toxicity was infrequent with an infection rate of 9% and no bleeding observed
 - Patients on the 21-day cycle had substantially more grade 3 and 4 TEAEs than patients on the 28-day cycle
- Including patients with SD, a large proportion of patients treated with melflufen had clinical benefit in O-12-M1[10]
 - Disease stabilization (≥SD), 76%
- Given the large proportion of patients who achieved disease stabilization, melflufen has potential to delay progressive disease (PD) and extend TTNT in patients with RRMM

Figure 1. Melflufen Mechanism of Action

Peptidase-enhanced activity in multiple myeloma cells

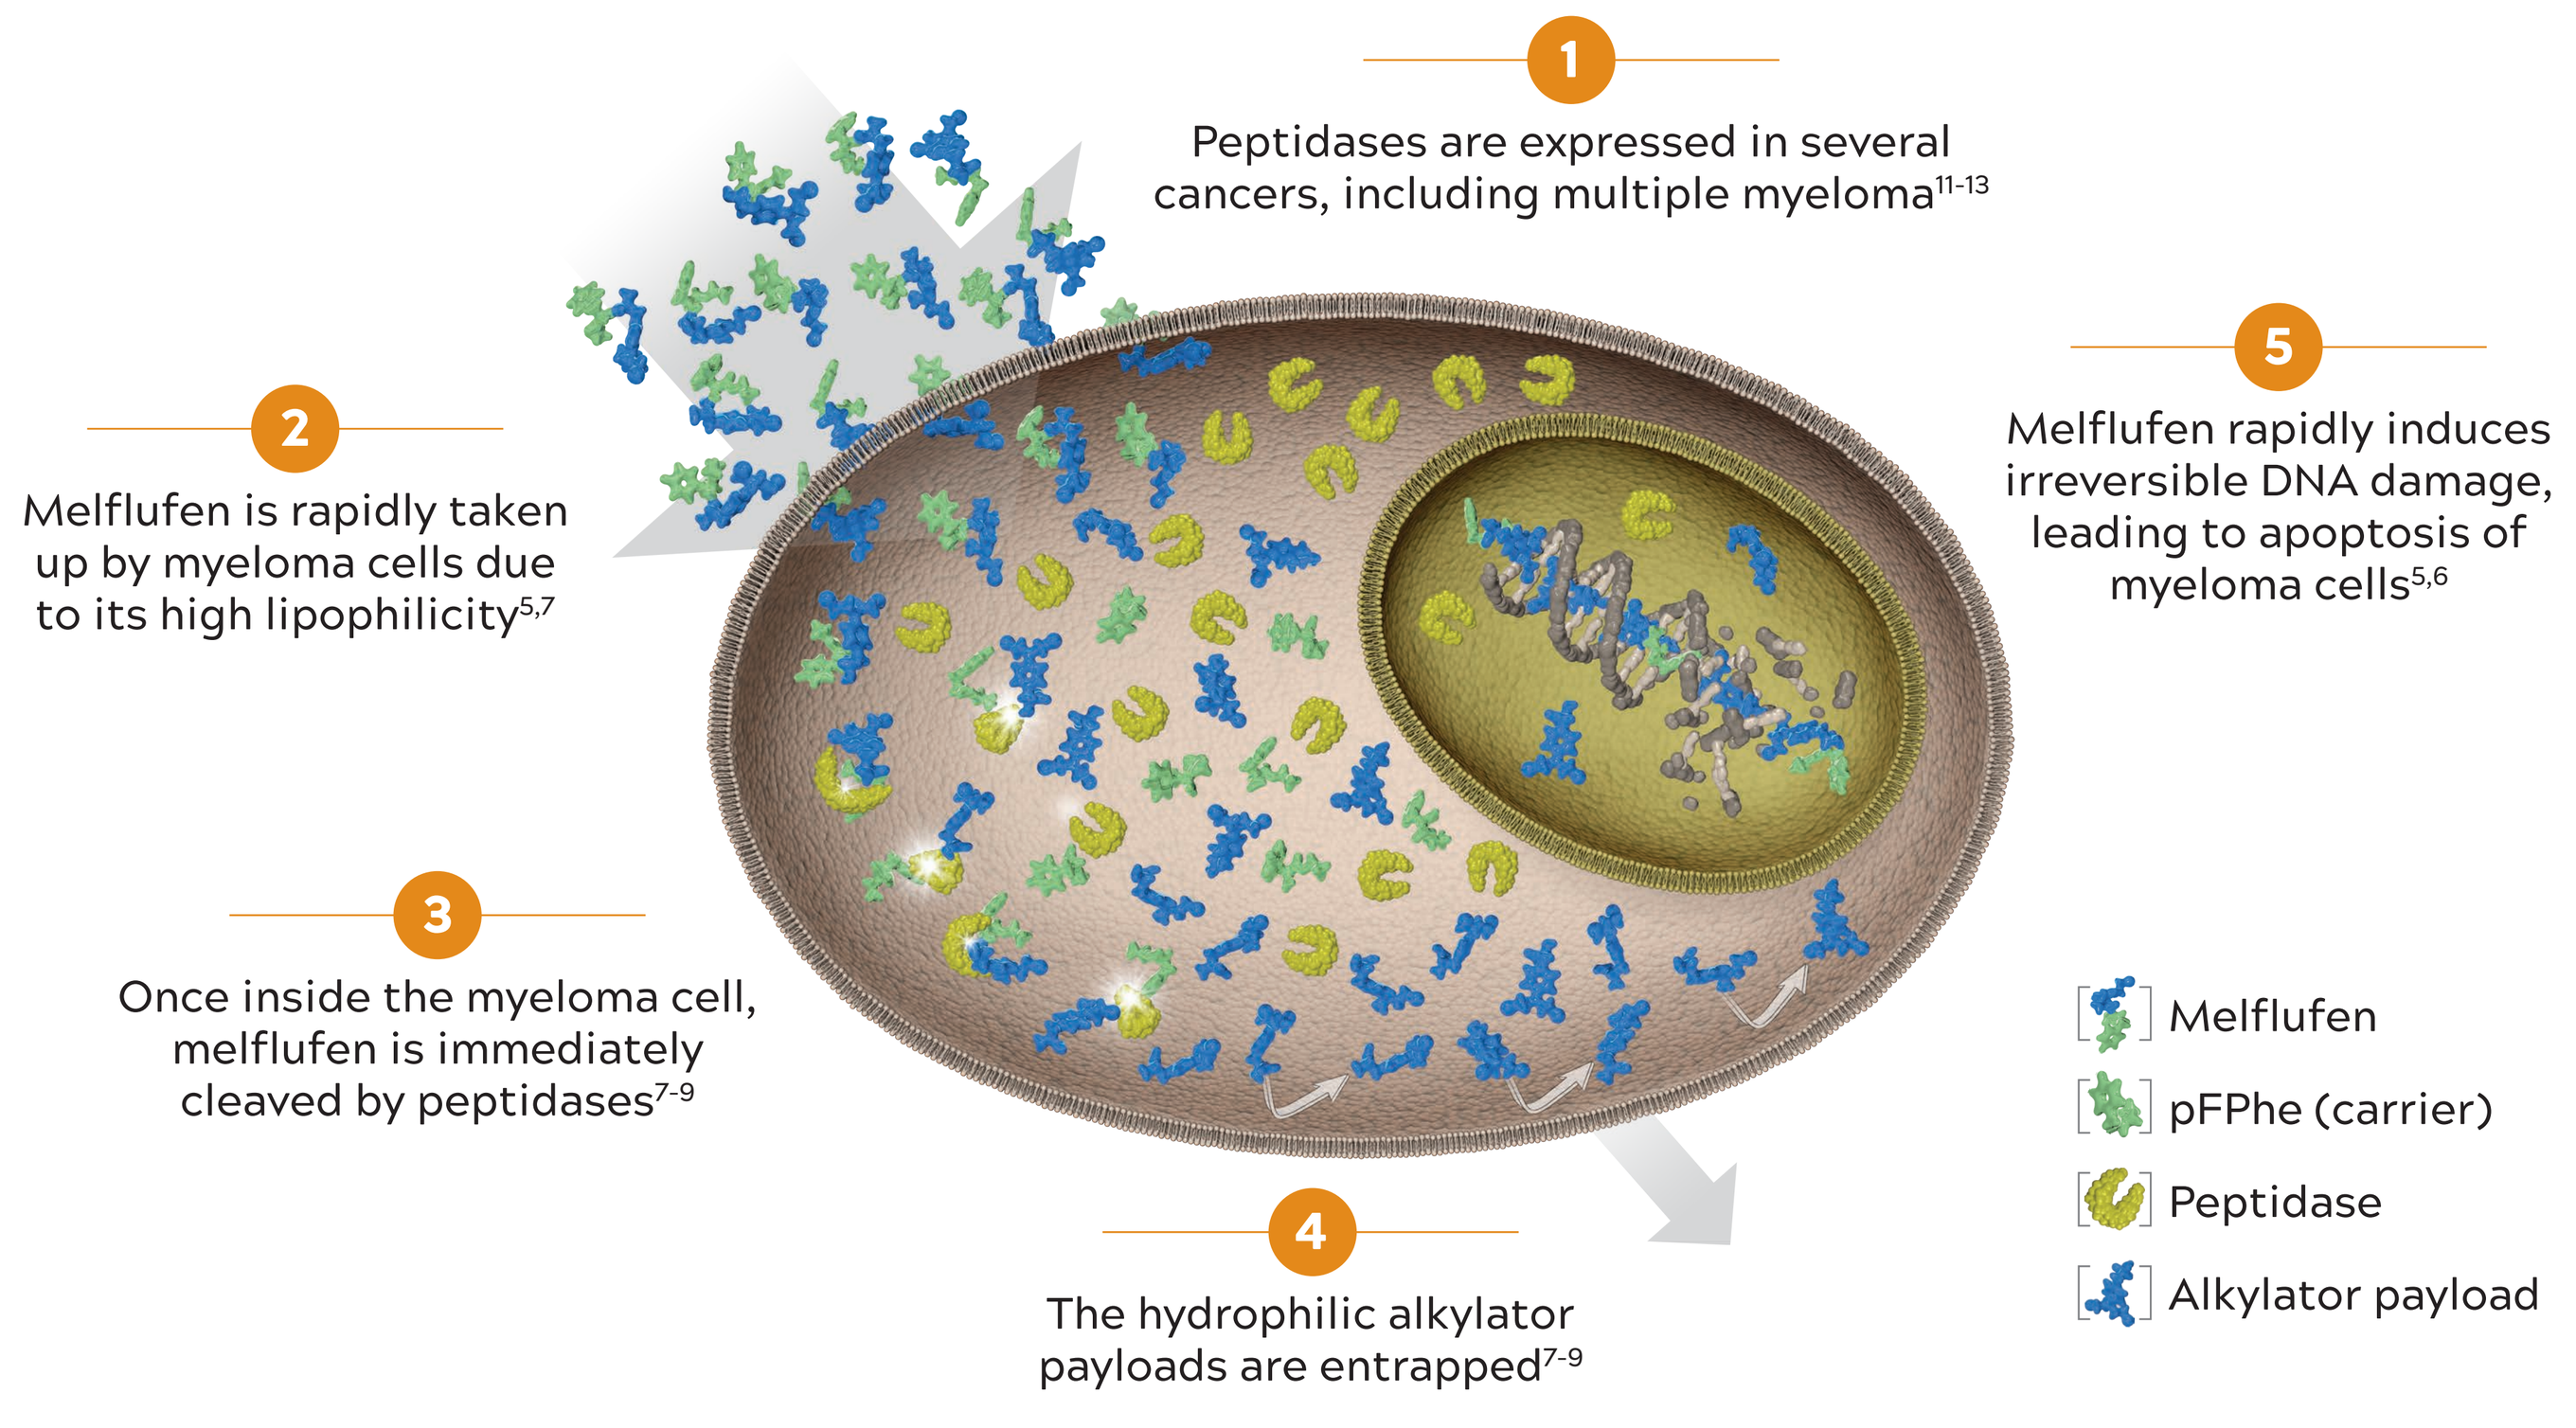

1
Peptidases are expressed in several cancers, including multiple myeloma[11-13]
2
Melflufen is rapidly taken up by myeloma cells due to its high lipophilicity[5,7]
3
Once inside the myeloma cell, melflufen is immediately cleaved by peptidases[7-9]
4
The hydrophilic alkylator payloads are entrapped[7-9]
5
Melflufen rapidly induces irreversible DNA damage, leading to apoptosis of myeloma cells[5,6]
Melflufen
pFPhe (carrier)
Peptidase
Alkylator payload

Melflufen is 50-fold more potent than melphalan in myeloma cells in vitro due to increased intracellular alkylator activity[5,7]

pFPhe, para-fluoro-L-phenylalanine.

OBJECTIVES

- To assess TTNT with melflufen plus dexamethasone in patients with RRMM in an exploratory, post hoc analysis of the phase 2 part of the O-12-M1 study
- To describe subsequent treatments in an RRMM patient population with a median of 4 to 5 prior lines of therapy

METHODS

Figure 2. Phase 2 O-12-M1 Study Design (NCT01897714)

N=45
Patients with RRMM and ≥2 prior lines of therapy including lenalidomide and bortezomib and PD on or within 60 days of completion of last therapy

→ **Melflufen + dexamethasone**: 21-Day cycle (n=28) | 28-Day cycle (n=17)

→ After PD or start of subsequent therapy, patients were followed for survival every 3 months for up to 24 months

Primary endpoint: ORR
Secondary endpoints: PFS, OS, safety

PFS, progression-free survival; PD, progressive disease; ORR, overall response rate; OS, overall survival; RRMM, relapsed/refractory multiple myeloma.

- Melflufen 40 mg was administered intravenously on day 1 of each 21- or 28-day cycle with oral dexamethasone 40 mg given weekly for up to 8 cycles or longer at the discretion of the investigator and sponsor
- Response was assessed by the investigator at each cycle by International Myeloma Working Group criteria
- TTNT was retrospectively reviewed and was defined in line with guidelines as time from start of melflufen plus dexamethasone to first subsequent therapy or death, whichever occurred first. An analysis of TTNT, in which deaths were censored, was performed to allow comparison with historical data (where both methods have been used)

RESULTS

PATIENT DISPOSITION

- As of 9 November 2017, 45 patients were treated in O-12-M1
- At data cutoff, 44 patients (98%) had discontinued melflufen plus dexamethasone (**Table 1**)
- Reasons for discontinuation were significantly different for patients on the 21-day cycle than on the 28-day cycle
- In total, 27 out of 41 patients (66%) received subsequent therapy (4 patients were still progression free at the time of data cutoff), which is in line with previously reported data in advanced RRMM in which 39% to 72% of patients received subsequent therapy[14-16]
- 9 Patients who discontinued because of AEs had grade 4 thrombocytopenia after the last cycle of melflufen (all in the 21-day cycle length group), with a median PFS of 9.3 months (95% CI, 6.5-not calculable [NC]), median DOR of 9.0 months (95% CI, 3.0-NC), and a median time between progression and start of subsequent therapy of 6 days
 - 8 out of the 9 patients (89%) received subsequent therapy

Table 1. Patient Disposition (and Supplemental Safety Information)

Disposition, n (%)	ITT (N=45)	21-Day Cycle (n=28)	28-Day Cycle (n=17)
Discontinued treatment	44 (98)	28 (100)	16 (94)
Reason for discontinuation			
AE[a]	18 (40)	16 (57)	2 (12)
PD	14 (31)	7 (25)	7 (41)
Lack of response	1 (2)	0	1 (6)
Death	2 (4)	0	2 (12)
Completed 8 cycles	9 (20)	5 (18)	4 (24)
Ongoing treatment	1 (2)	0	1 (6)
Grade 3/4 thrombocytopenia[b]	28 (62)	24 (86)	4 (24)

AE, adverse event; ITT, intention-to-treat; PD, progressive disease.
[a]Discontinuation rate due to AE was high in the 21-day cycle group due to thrombocytopenia. Please see supplemental safety information.
[b]Supplemental safety information.

Table 2. Baseline Patient Characteristics for the ITT Population and by Dosing Cycle Length

Characteristic	ITT (N=45)	21-Day Cycle (n=28)	28-Day Cycle (n=17)
Median age (range), years	66 (47-78)	64 (48-74)	70 (47-78)
Gender (male/female), %	67/33	64/36	71/29
ISS stage at study entry (I/II/III), %	33/40/20	32/43/21	35/35/18
Median no. prior lines (range)	4 (2-14)	4 (2-14)	4 (2-14)
Albumin level at study entry			
≥3.5 g/dL, n (%)	31 (69)	18 (64)	13 (76)
<3.5 g/dL, n (%)	14 (31)	10 (36)	4 (24)
High-risk cytogenetics,[a] n (%)	20 (44)	12 (43)	8 (47)
High LDH (1.5×ULN), n (%)	23 (51)	15 (54)	8 (47)
Single refractory,[b] n (%)	41 (91)	25 (89)	16 (94)
Double refractory,[c] n (%)	30 (67)	18 (64)	12 (71)
Triple refractory,[d] n (%)	3 (7)	2 (7)	1 (6)
Alkylator refractory,[e] n (%)	24 (53)	16 (57)	8 (47)

ISS, International Staging System; ITT, intention-to-treat; LDH, lactate dehydrogenase; PI, proteasome inhibitor; ULN, upper limit of normal.
[a]Defined as del(17p), t(4;14), t(14;16), t(14;20), or gain(1q). [b]At least 1 PI or IMiD. [c]At least 1 PI and IMiD. [d]At least 1 PI and IMiD and daratumumab
[e]Melphalan, cyclophosphamide, or bendamustine.

Figure 3. TTNT and PFS in the ITT Population (N=45)

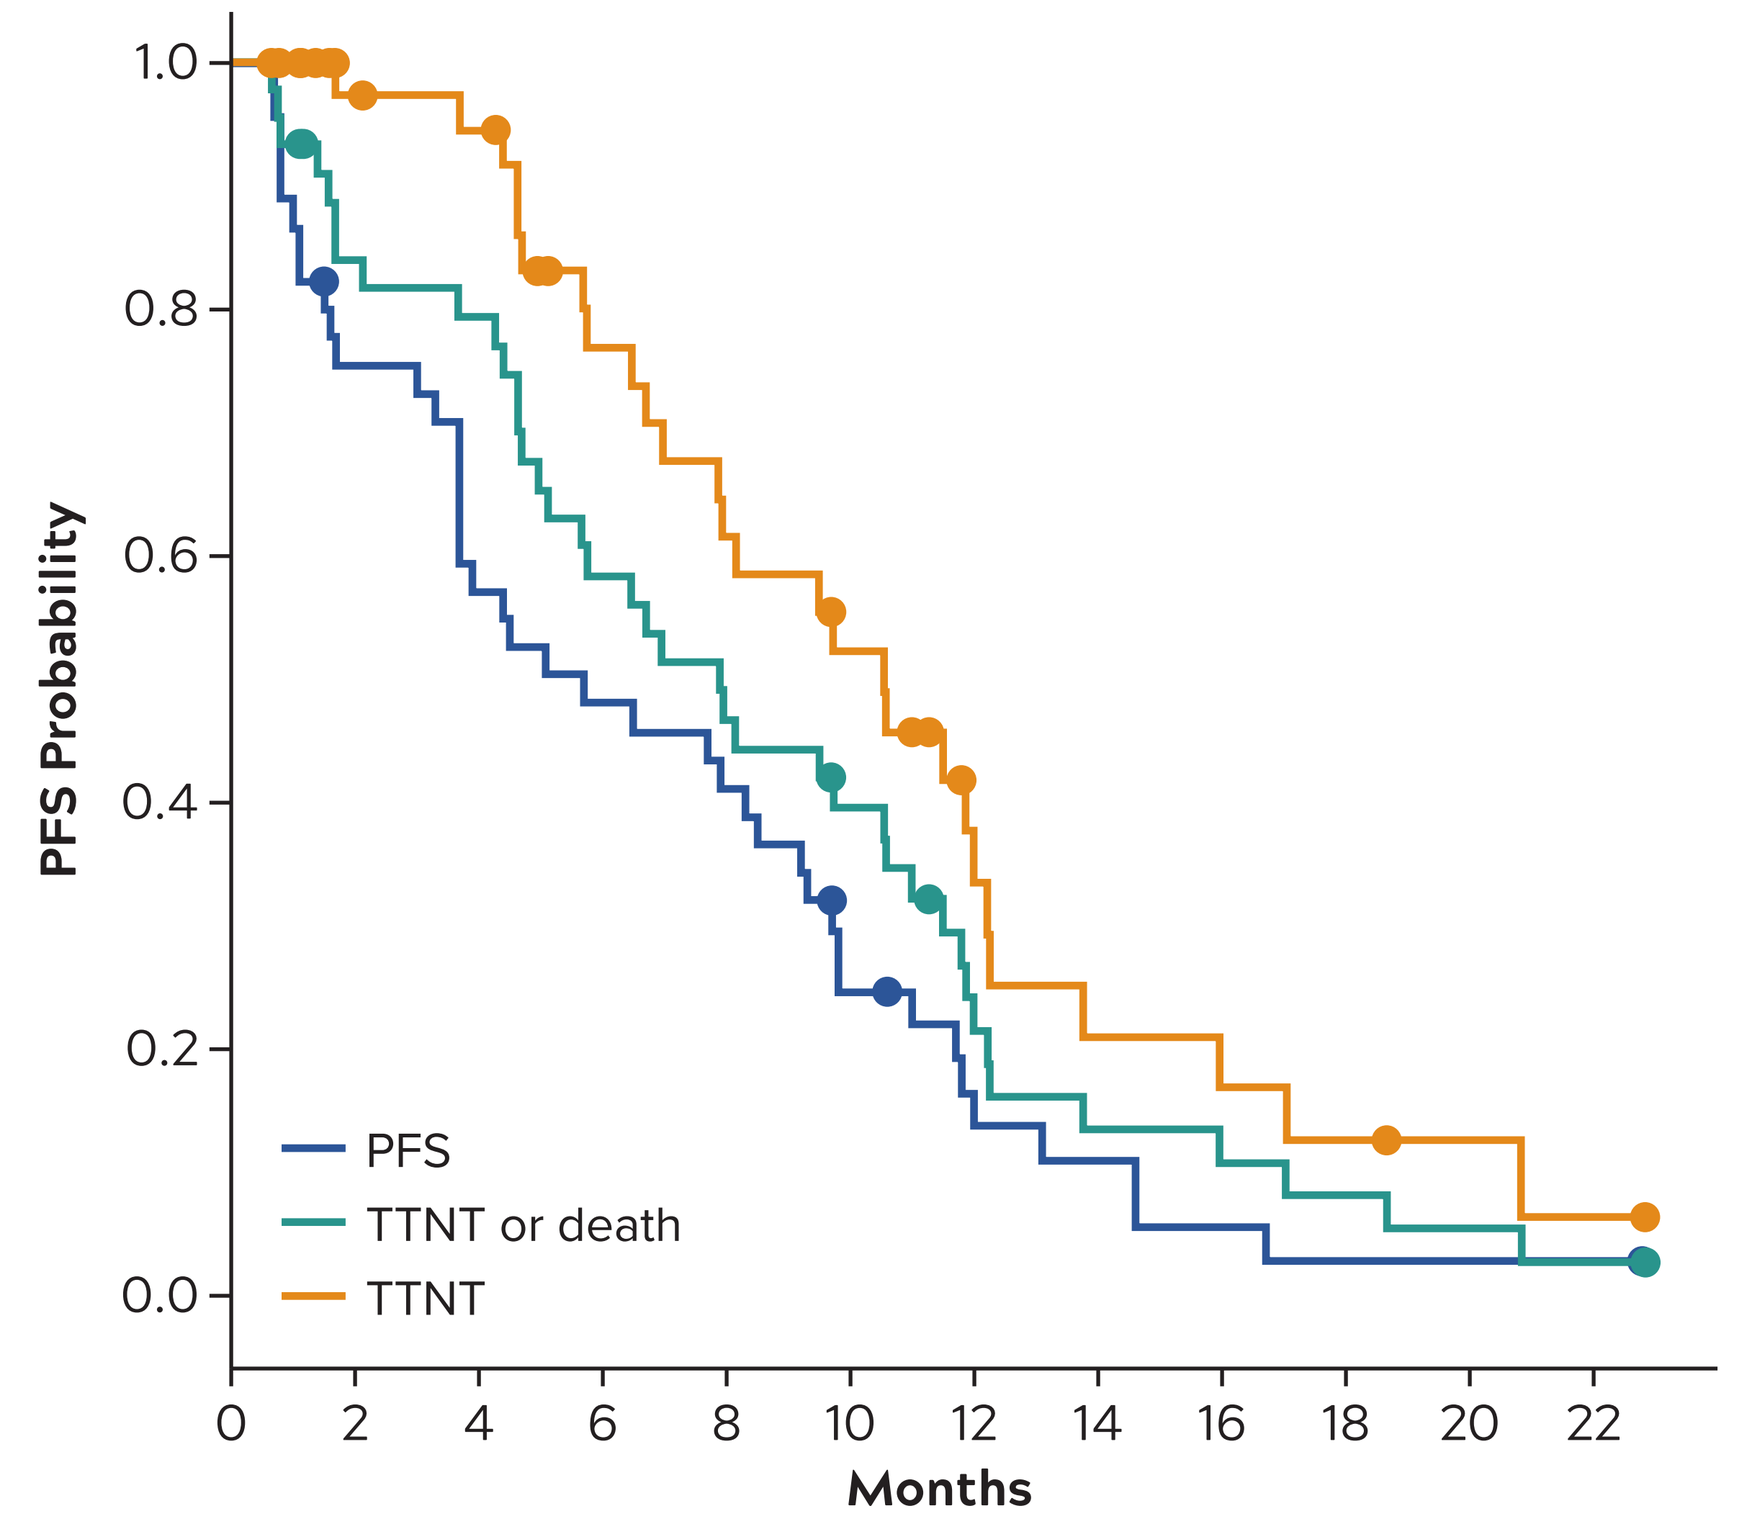

PFS Probability
1.0
0.8
0.6
0.4
0.2
0.0
PFS
TTNT or death
TTNT
0 2 4 6 8 10 12 14 16 18 20 22
Months

An event was defined as subsequent treatment or death for the TTNT analysis (in the auxiliary analysis death was instead censored).
An event was defined as PD or death, whichever occurred first for the PFS analysis.
ITT, intention-to-treat; PD, progressive disease; PFS, progression-free survival; TTNT, time to next treatment.

- Median TTNT was 7.9 months (95% CI, 5.68-11.01), with 40 events in 45 patients (**Figure 3**)
- Median TTNT when censoring for deaths was 10.6 months (95% CI, 8.0-12.3; **Figure 3**)
- Median PFS was 5.7 months (95% CI, 3.7-9.3), with 41 events in 45 patients (**Figure 3**)

Figure 4. OS in the ITT Population (N=45)

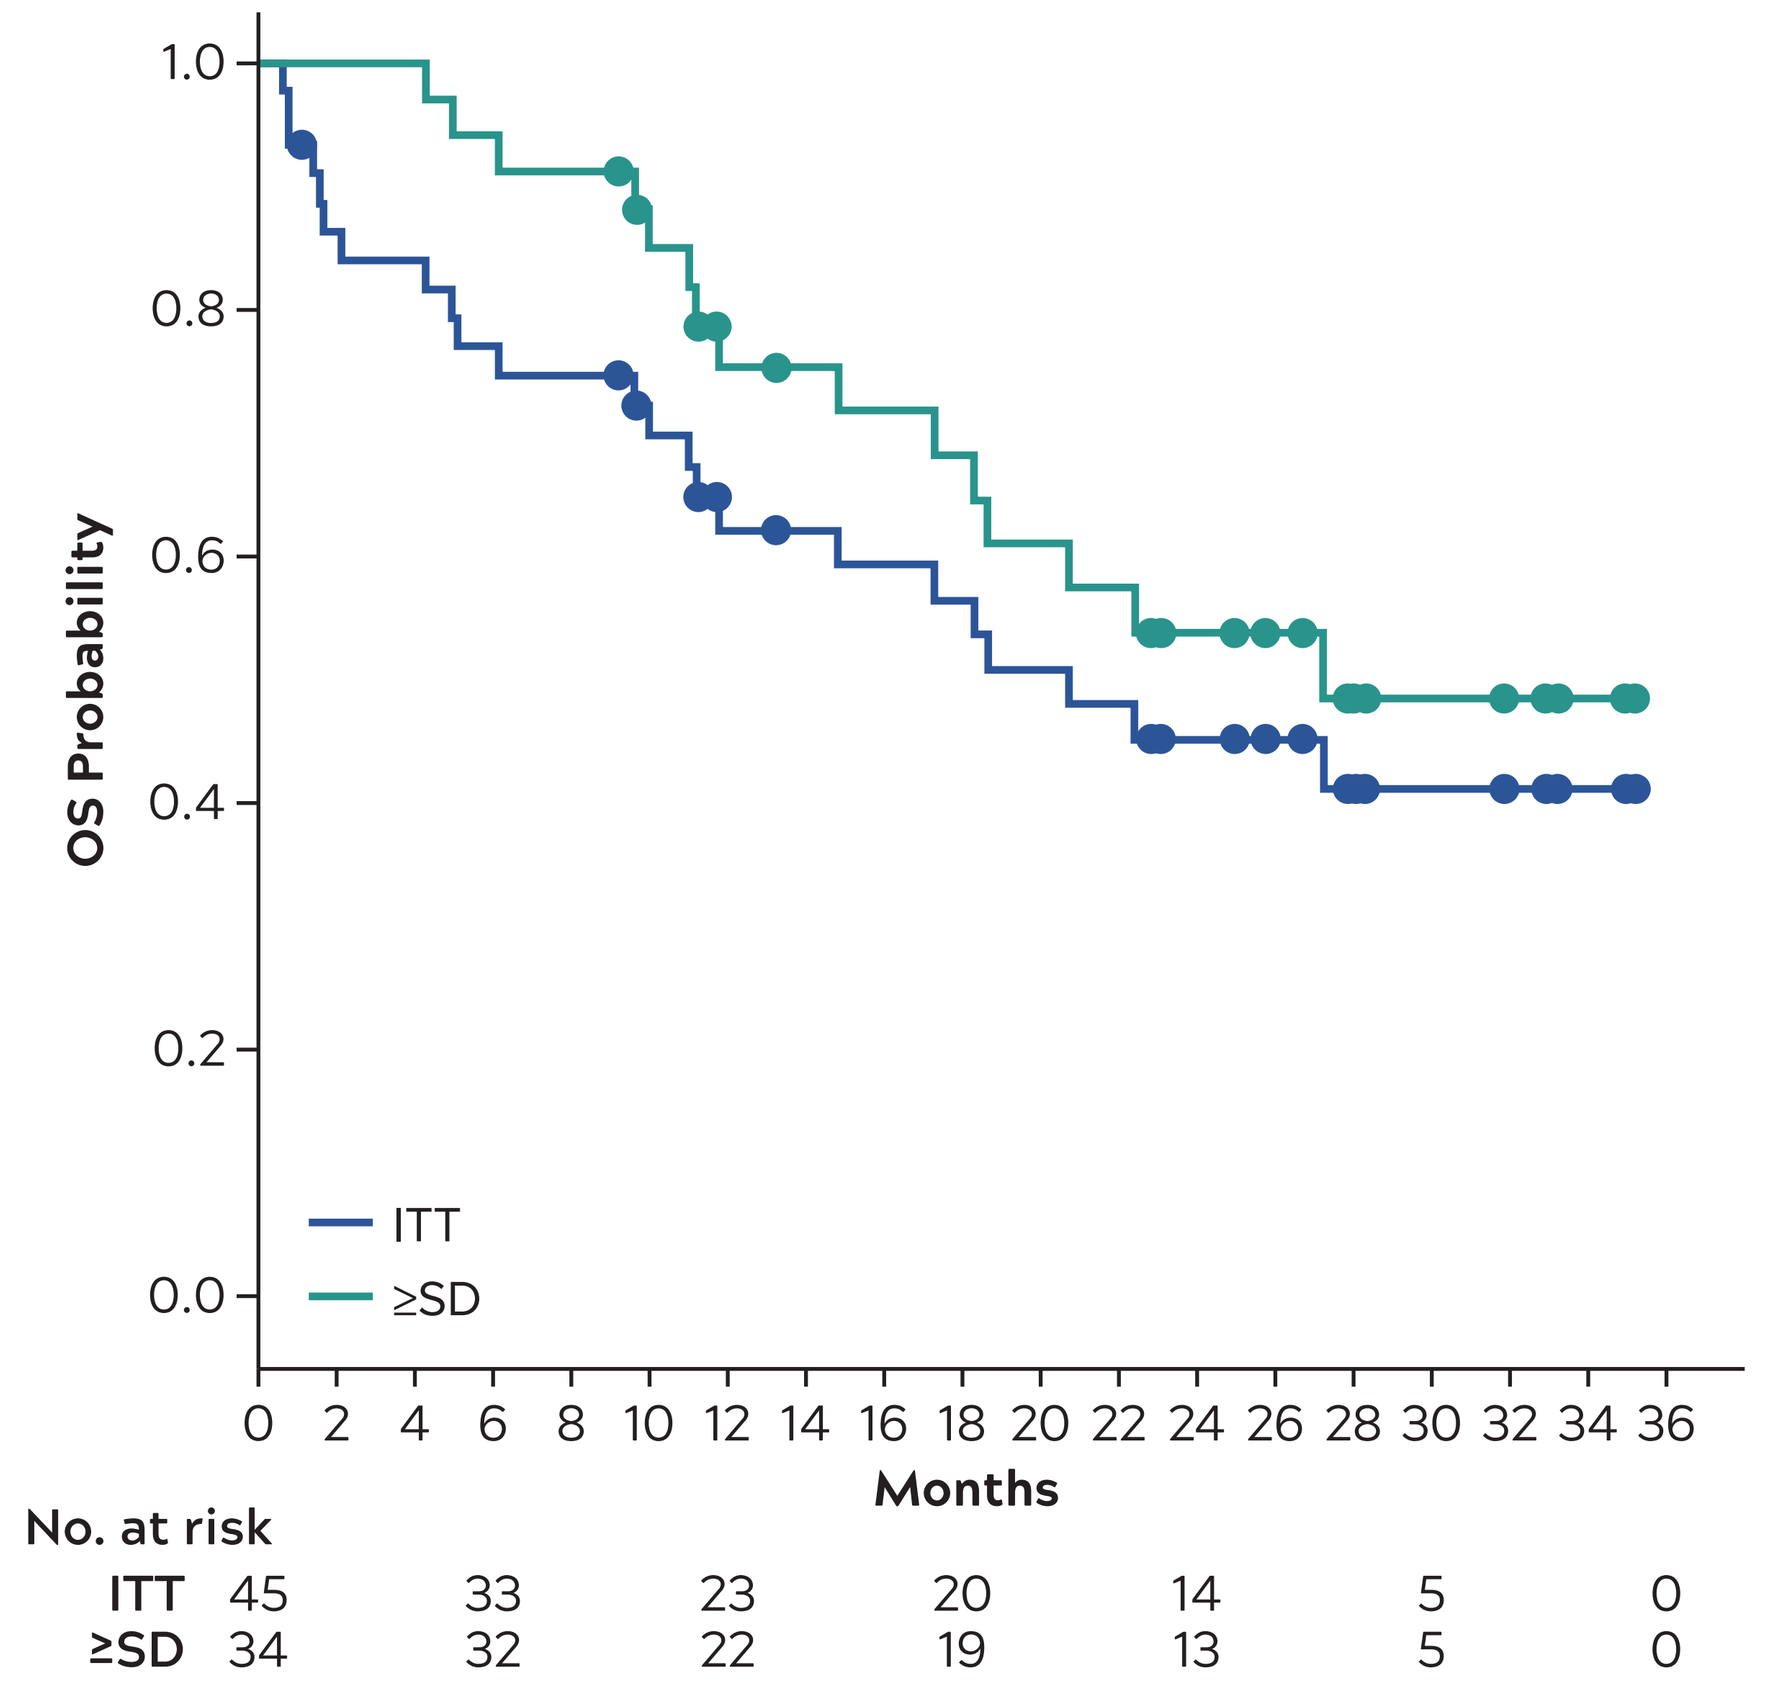

OS Probability
1.0
0.8
0.6
0.4
0.2
0.0
ITT
≥SD
0 2 4 6 8 10 12 14 16 18 20 22 24 26 28 30 32 34 36
Months
No. at risk
ITT 45 33 23 20 14 5 0
≥SD 34 32 22 19 13 5 0

An event was defined as death for the OS analysis.
ITT, intention-to-treat; OS, overall survival; SD, stable disease.

- Median OS in the total population was 20.7 months (95% CI, 11.8-NC), with 23 events in 45 patients, and 27.2 months (95%CI, 18.3-NC) in patients with SD or better (**Figure 4**)

Table 3. TTNT With Melflufen in O-12-M1 and Other Agents in RRMM

Agent or Regimen	No. of Patients	Prior Lines of Therapy	Death as an Event or Censored	Median TTNT, mo
Melflufen	45	4	Censored	10.6
Pomalidomide or carfilzomib[17]	Pomalidomide: 264 Carfilzomib: 190	2+	Censored	Pomalidomide: 11.9 Carfilzomib: 9.4
Melflufen	45	4	Event	7.9
VRd or KRd[18]	VRd: 343 KRd: 139	1-3	Event	VRd: 12.9 KRd: 8.7
Daratumumab[19]	126	4	Event	5.9

KRd, carfilzomib + lenalidomide + dexamethasone; RRMM, relapsed/refractory multiple myeloma; TTNT, time to next treatment; VRd, bortezomib + lenalidomide + dexamethasone.

- The data presented suggest a TTNT with melflufen plus dexamethasone that is as good as or better than that of other RWE studies of agents in RRMM (**Table 3**)

SUBSEQUENT THERAPY

- In total, 66% of the patients went on to receive subsequent therapy (27/41 patients, with 4 patients still progression free at the time of the data cutoff)
- Variability in subsequent therapy was high, indicating a high unmet medical need in this patient population
- The majority of patients (52%) received single agent with or without steroid therapy, and approximately half of the patients receiving subsequent therapy (44%) received ≥2 subsequent lines of therapy

CONCLUSIONS AND FUTURE DIRECTIONS

- Melflufen plus dexamethasone treatment results in disease stabilization (≥SD) in 76% of patients, which translates to a median TTNT of 7.9 months (10.6 months when censoring at time of death) in heavily pretreated patients with RRMM, which compares favorably with other relevant trials
- The median OS of 20.7 months in this advanced RRMM population suggests that melflufen therapy is associated with a long-term benefit, allowing patients to receive further treatment to control disease
- Longer TTNT for patients with RRMM is associated with clinical benefit as well as health economic value for payors. The reported ranges of median PFS/TTNT values in real-world are generally shorter than those in phase 3 clinical studies.[20] Real world data will be gathered for melflufen in future studies
- Results support those of previous reports showing the promising efficacy profile of melflufen for the treatment of RRMM
- Furthermore, data from O-12-M1 suggest that, in addition to the established clinical benefit from ≥PR and ≥MR responses, SD is also a clinically meaningful response in patients with RRMM treated with melflufen plus dexamethasone and that such patients should continue to stay on treatment. This will be further investigated in ongoing and future studies
- The high variability in subsequent therapies after melflufen plus dexamethasone indicates a lack of good treatment options and a significant unmet medical need in patients with advanced RRMM
- OP-103 OCEAN is an ongoing phase 3, randomized, global study that is further evaluating the efficacy and safety of melflufen plus dexamethasone versus pomalidomide plus dexamethasone in patients with RRMM refractory to lenalidomide (NCT03151811)

REFERENCES

1. Chen CC, et al. *J Manag Care Spec Pharm*. 2017;23:236-246.
2. Yong K, et al. *Br J Haematol*. 2016;175:252-264.
3. Van Beurden-Tan CHY, et al. *Blood*. 2016;128(suppl, abstr):2141.
4. Arikian SR, et al. *Blood*. 2015;126(suppl, abstr):3294.
5. Chauhan D, et al. *Clin Cancer Res*. 2013;19:3019-3031.
6. Ray A, et al. *Br J Haematol*. 2016;174:397-409.
7. Wickström M, et al. *Oncotarget*. 2017;8:66641-66655.
8. Wickström M, et al. *Biochem Pharmacol*. 2010;79:1281-1290.
9. Gullbo J, et al. *J Drug Target*. 2003;11:355-363.
10. Richardson PG, et al. *Blood*. 2017;130(suppl, abstr):3150.
11. Hitzerd SM, et al. *Amino Acids*. 2014;46:793-808.
12. Moore HE, et al. *Mol Cancer Ther*. 2009;8:762-770.
13. Wickström M, et al. *Cancer Sci*. 2011;102:501-508.
14. Kumar S, et al. *Blood*. 2009;114(suppl, abstr):2878.
15. Gandhi UH, et al. *Blood*. 2018;132(suppl, abstr):3233.
16. Single Technology Appraisal. Daratumumab monotherapy for treating relapsed and refractory multiple myeloma [ID933]. https://www.nice.org.uk/guidance/ta510/documents/committee-papers (p218). Accessed May 21, 2019.
17. Maciocia N, et al. *Clin Lymphoma Myeloma Leuk*. 2015;15(suppl, abstr):PO-359.
18. Chari A, et al. *Blood*. 2017;130(suppl, abstr):1818.
19. Lakshman A, et al. *Am J Hematol*. 2017;92(11):1146-1155.Insert reference 20 below this:
20. Richardson PG, et al. *Blood Cancer J*. 2018;8:109.

ACKNOWLEDGMENTS

The authors would like to thank the patients who volunteered to participate in the study, the staff and the study sites who cared for them, and the CRO involved in data gathering and analyses.
Medical writing support was provided by Jennifer Leslie, PhD, of Team 9 Science with funding from Oncopeptides.

DISCLOSURES

SB: honoraria from Celgene, Janssen-Cilag, Amgen, and Bristol-Myers Squibb; consulting/advisory role with Celgene, Janssen-Cilag, Amgen, Takeda, and Karyopharm. **PGR:** consulting/advisory role for Oncopeptides. **PV:** consulting/advisory role for Adaptive Biotechnologies, Bristol-Myers Squibb, Celgene, Janssen, Novartis, Oncopeptides, Takeda, and TeneoBio. **TP:** consulting/advisory role for Janssen, Celgene, Takeda, AbbVie, and Genmab; research funding from Janssen. **UHM:** honoraria from Janssen, Oncopeptides, Celgene, Takeda, and Amgen; consulting/advisory role for Amgen. **JAZ:** honoraria from Bristol-Myers Squibb and Celgene; consulting/advisory role with Alnylam, Prothena, Amgen, Takeda, Celgene, Caelum, and Oncopeptides; research funding from Bristol-Myers Squibb and Celgene; travel/accommodations/expenses from Bristol-Myers Squibb, Celgene, Alynlam, Takeda, Amgen, Oncopeptides, and Prothena. **BR:** employment with Select Specialty Hospital; consulting/advisory role with Seattle Genetics; speakers' bureau for Celgene; and research funding from Janssen, Amgen, GlaxoSmithKline, Oncopeptides, and Celgene. **SZ, JH, JO:** employment and equity ownership with Oncopeptides. **PS:** honoraria and consulting/advisory role for Amgen, Celgene, Janssen, Seattle Genetics; research funding from Janssen and Celgene.

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ASCO® and the author of this poster.

Presented at the 2019 annual meeting of the American Society of Clinical Oncology (ASCO), May 31 - June 4, 2019; Chicago, IL